EP0031 a next-generation selective RET inhibitor (SRI): Correlation of molecular and clinical responses in patients with RET alteration positive solid tumours naïve to or following prior SRI

1295P

Elena Garralda[1], Judy S. Wang[2], Andrew G. Gianoukakis[3], Matthew H. Taylor[4], Daniel Morgensztern[5], Susanne M. Arnold[6], David Spigel[7], Stephen Liu[8], Matthew G. Krebs[9], Martin Forster[10], Javier Garcia-Corbacho[11], Luis Paz Ares[12], Liz Clark[13], Geoff Fisher[13], Sue Brook[13], Sital Patel[13], Tobias Arkenau[13], Brooke Overstreet[14], Geethika Yalamanchili[14], Pilar Garrido[15]

1. Hospital Universitari Vall D'Hebron and Institute of Oncology, Barcelona, Spain 2. Florida Cancer Specialists/Sarah Cannon Research Institute, Sarasota, Florida, USA 3. The Lundquist Institute at Harbor-UCLA Medical Center, Torrance, and The David Geffen School of Medicine at UCLA, Los Angeles, California, USA 4. Providence Cancer Institute, Portland, Oregon, USA 5. Washington University, St Louis, Missouri, USA 6. University of Kentucky/Markey Cancer Center, Lexington, Kentucky, USA 7. Sarah Cannon Research Institute, Tennessee, USA 8. Lombardi Comprehensive Cancer Center, Georgetown University, Washington, DC, USA 9. The University of Manchester and The Christie NHS Foundation Trust, Manchester, UK 10. University College London Hospital, London, UK 11. Hospital Universitario Virgen de la Victoria/IBIMA, Malaga, Spain 12. Hospital Universitario 12 de Octubre, Madrid, Spain 13. Ellipses Pharma, London, UK 14. Guardant Health, Redwood City, California, USA 15. Hospital Universitario Ramón y Cajal, Madrid, Spain

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without including written permission of the authors.

BACKGROUND

- EP0031 (A400/KL590586) is a next gen selective RET inhibitor (SRI) with greater potency against common RET alterations including resistance mutations, greater brain penetration, and produces deeper responses, compared to 1st gen SRIs in preclinical NSCLC models[1]
- A Phase 1/2 study in China (KL400-I/II-01, NCT05265091), reported a high ORR in patients with RET fusion positive NSCLC naïve to SRI, responses in patients with prior 1st gen SRI, activity against brain metastases, and encouraging tolerability[1]. Two registration cohorts are still ongoing
- Ellipses Pharma recently reported Phase 1 dose escalation and expansion data from the first study of EP0031-101 (NCT05443126) in patients in the US and Europe, at ASCO 2024[2]
- EP0031 has been granted FDA Orphan Drug (ODD) and Fast Track Designation
- Globally, over 370 patients and 84 healthy volunteers have received EP0031

STUDY DESIGN

Dose escalation

- Dose escalation was based on a rolling 6 design, with patients dosed at levels ranging from 20 to 120 mg QD in 28-day cycles. The 60, 90 and 120 mg cohorts were expanded to a total of 40 patients

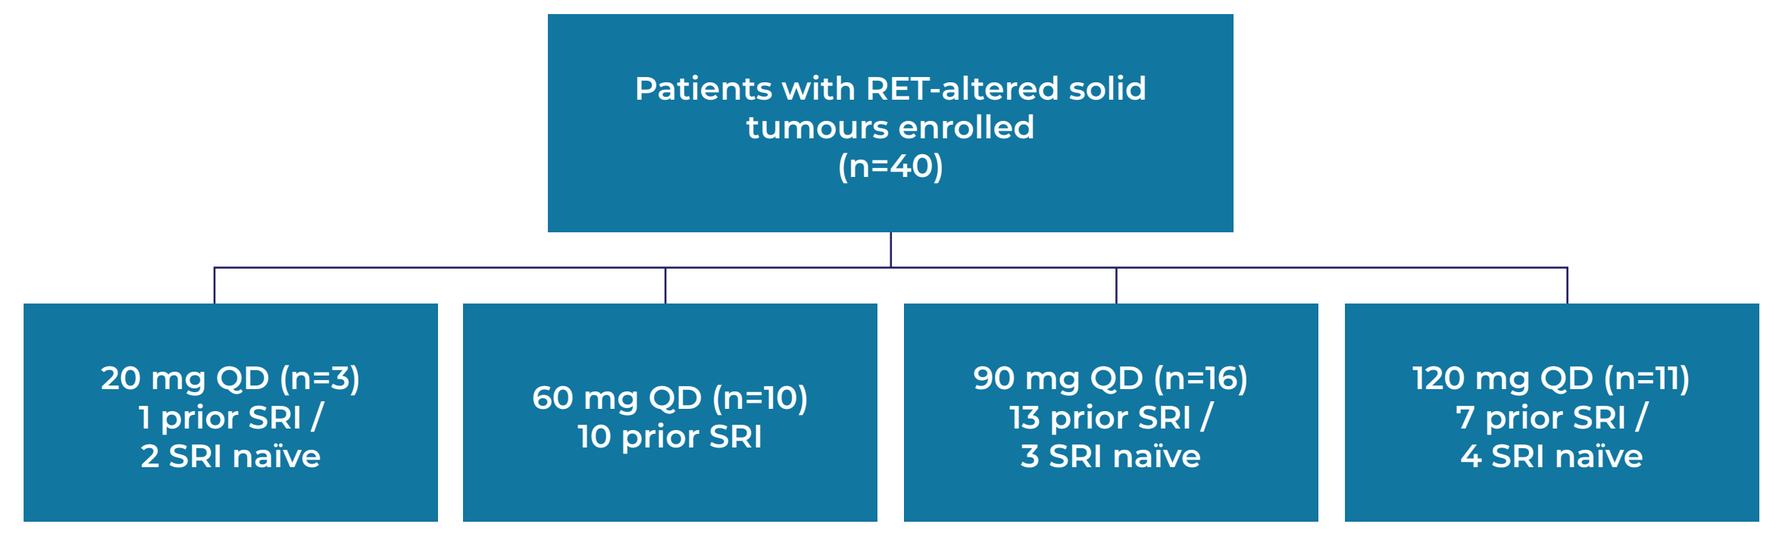

Patients with RET-altered solid tumours enrolled (n=40)
20 mg QD (n=3) 1 prior SRI / 2 SRI naïve
60 mg QD (n=10) 10 prior SRI
90 mg QD (n=16) 13 prior SRI / 3 SRI naïve
120 mg QD (n=11) 7 prior SRI / 4 SRI naïve

Figure 1. EP0031 Monotherapy Dose Escalation Study Schema

Eligibility

- Patients with RET-altered NSCLC, medullary thyroid cancer (MTC) or other solid tumours (measurable or non-measurable disease); ≥ 18 years of age, PS 0 or 1, with/without asymptomatic, stable brain metastases

Key endpoints

- Safety and tolerability
- Determine MTD/or RP2D
- PK, biomarkers (ctDNA)
- ORR, BOR, DOR, change in tumour size, PFS, OS

Exploratory endpoints

- PD responses and EP0031 mode of action
- Relationship between PK exposure and PD biomarkers
- Predictors of response and resistance

Biomarker methodology

- Plasma samples were collected at baseline, on-treatment, and end of treatment (EOT) (Figure 2) and analysed using Guardant Infinity, a validated NGS ctDNA assay[3]

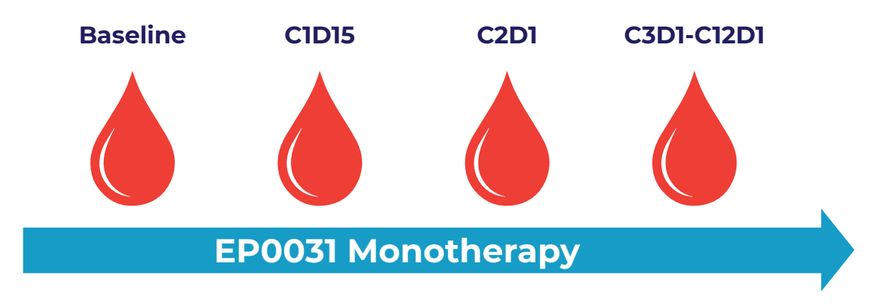

Baseline
C1D15
C2D1
C3D1-C12D1
EP0031 Monotherapy

Figure 2. ctDNA blood draws

- ctDNA (circulating tumour DNA) are fragments of tumour cells or tumour originating cell-free DNA (cfDNA) circulating in the blood stream
- Tumour Fraction (TF) is the ctDNA (tumour originating cell-free DNA) as a proportion of cfDNA circulating in the blood stream
- Percentage ctDNA change assessed by genomic variant and methylation-based methods
- Methylation Tumour Fraction (mTF) quantified by measuring cancer-type specific differentially methylated regions (DMRs), as previously described (DMRs are regions determined to be methylated in cancer patients and not in cancer-free donors)[4]

RESULTS

Summary of demography and patient characteristics

A total of 40 patients were enrolled across dose cohorts of 20mg (N=3), 60mg (N=10), 90mg (N=16) and 120 mg (N=11). At the data cut-off (11 June 2024):

- 40 patients were evaluable for safety: 22 NSCLC, 12 MTC, and 6 patients with other tumour types; 23 female, 17 male; median age 58 years (32-77); 18 ECOG 0, 22 ECOG 1
- 33 patients (25 with prior SRI) RECIST evaluable for efficacy: 16 NSCLC, 12 MTC, 1 neuroendocrine CRC, 1 pancreatic, 1 endometrioid uterine carcinoma, 1 neuroendocrine lung, 1 papillary thyroid
- Patients with prior SRI treatment had received a median of 2.5 (range 1-6) prior lines of treatment

Safety and tolerability

Table 1 summarises safety findings by dose

- No dose-limiting toxicities or treatment-related deaths were reported
- The most common Grade 1 or 2 TRAEs were: headache, constipation, hyperphosphataemia, blurred vision, keratitis, ALT and AST increase, dry mouth. Other Grade 1 or 2 TEAEs were: dizziness, fatigue, creatinine increase, nausea, back pain, dyspnoea
- The most common Grade 3 TRAE were: headache (5%), AST and ALT increase (both 5%), hyponatraemia (5%), hypertension (5%), keratitis (2.5%)
- Most adverse events manageable with temporary dose interruptions (on average 7 days) and reductions, with one event of decreased visual acuity leading to treatment discontinuation

Table 1. Summary of safety findings by dose

	EP0031 Dose Level				
	20 mg (n=3)	60 mg (n=10)	90 mg (n=16)	120 mg (n=11)	Overall (n=40)[1]
TEAEs, all grades	3	9	16	11	39 (97.5%)
TEAEs, Grade ≥3	3	6	10	8	27 (67.5%)
TRAEs, all grades	3	7	14	10	34 (85.0%)
TRAEs, Grade ≥3	3	1	3	4	11 (27.5%)
TRAEs leading to Drug Interruption	1	1	6	7	15 (37.5%)
TRAEs leading to Dose Reduction	1	1	2	4	8 (20%)
TRAEs leading to Treatment Discontinuation	0	0	0	1	1 (2.5%)

AE, adverse event; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event
[1] Number of patients is used as denominator to calculate percentages. Note: Cut-off date: 11JUN2024

Efficacy and biomarkers

Figure 3. EP0031-101 Best % Change in Target Lesions

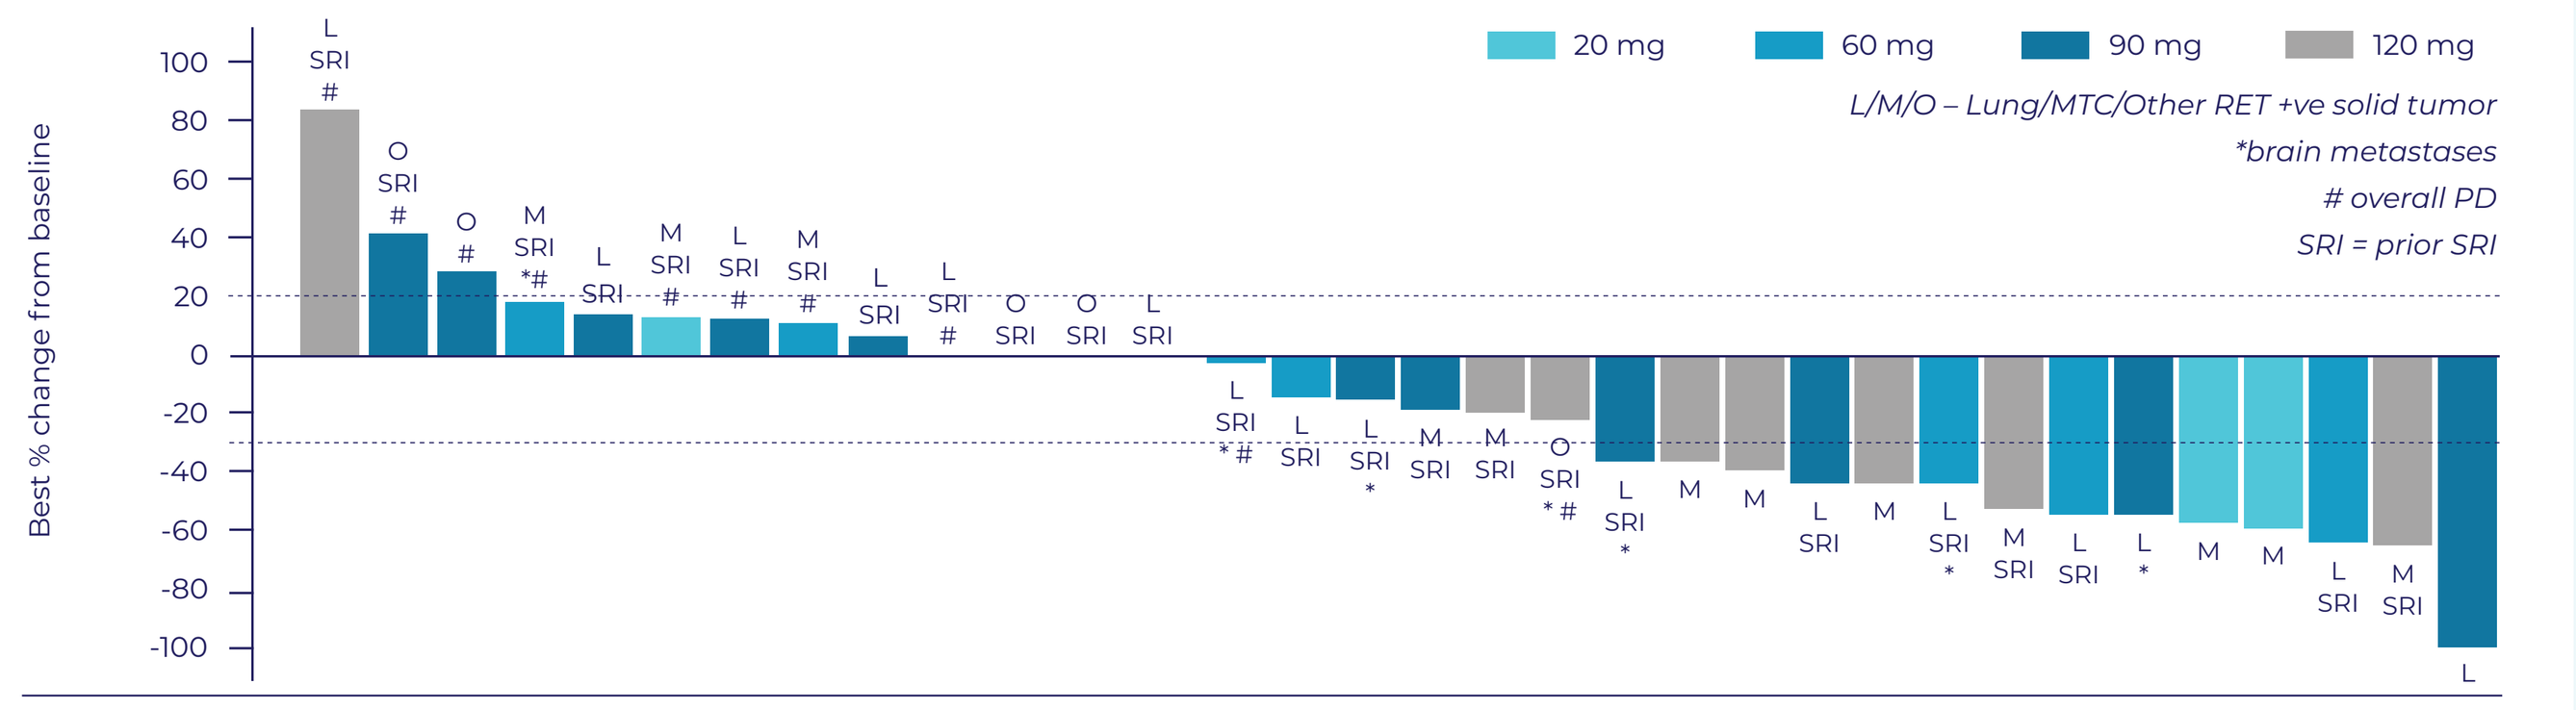

20 mg
60 mg
90 mg
120 mg
L/M/O – Lung/MTC/Other RET +ve solid tumor
*brain metastases
overall PD
SRI = prior SRI
Best % change from baseline

In the 33 RECIST response evaluable patients, EP0031 resulted in stabilisation of disease or marked tumour shrinkage and response across doses of 20-120mg:

Prior 1st gen SRI

- RET fusion +ve NSCLC: cPRs in 5 and SD in 5 out of 14 patients (at 60 and 90mg); DCR 71%
- Advanced MTC: 2 cPRs (at 120 mg) and 2 SD (at 90 and 120mg) out of 7 patients; DCR 57%
- Other solid tumours: 2 out of 4 patients SD (pancreatic and papillary thyroid at 60 mg); DCR 50%

SRI naïve

- RET fusion +ve NSCLC : CR and PR observed in 2 out of 2 patients (at 90mg)
- Advanced MTC: 4 out of 5 patients achieved a cPR, and 1 had uPR (at 120 mg)

NSCLC CNS responses

- NSCLC prior SRI: 3 out of 4 patients had complete resolution of brain lesions at 60 and 90mg
- NSCLC SRI naïve: 1 patient that presented with CNS disease had complete resolution of brain lesions at 90mg

Figure 4. 41 year-old female with KIF5B-RET fusion +ve NSCLC with L730V, L730I RET roof mutations who received prior pralsetinib [NSCLC-2]

Patient history

- Prior therapy: platinum-based doublet chemotherapy + IO, followed by pralsetinib for 18 months (BOR PR)
- Patient treated with EP0031, 60mg QD, escalated to 90mg QD after 5 cycles; cPR, CR in the brain
- Sustained reduction in methylation TF and ctDNA at C13D1 (-89%) compared to baseline (see adjacent figure)
- Patient continues to receive treatment after 16+ cycles

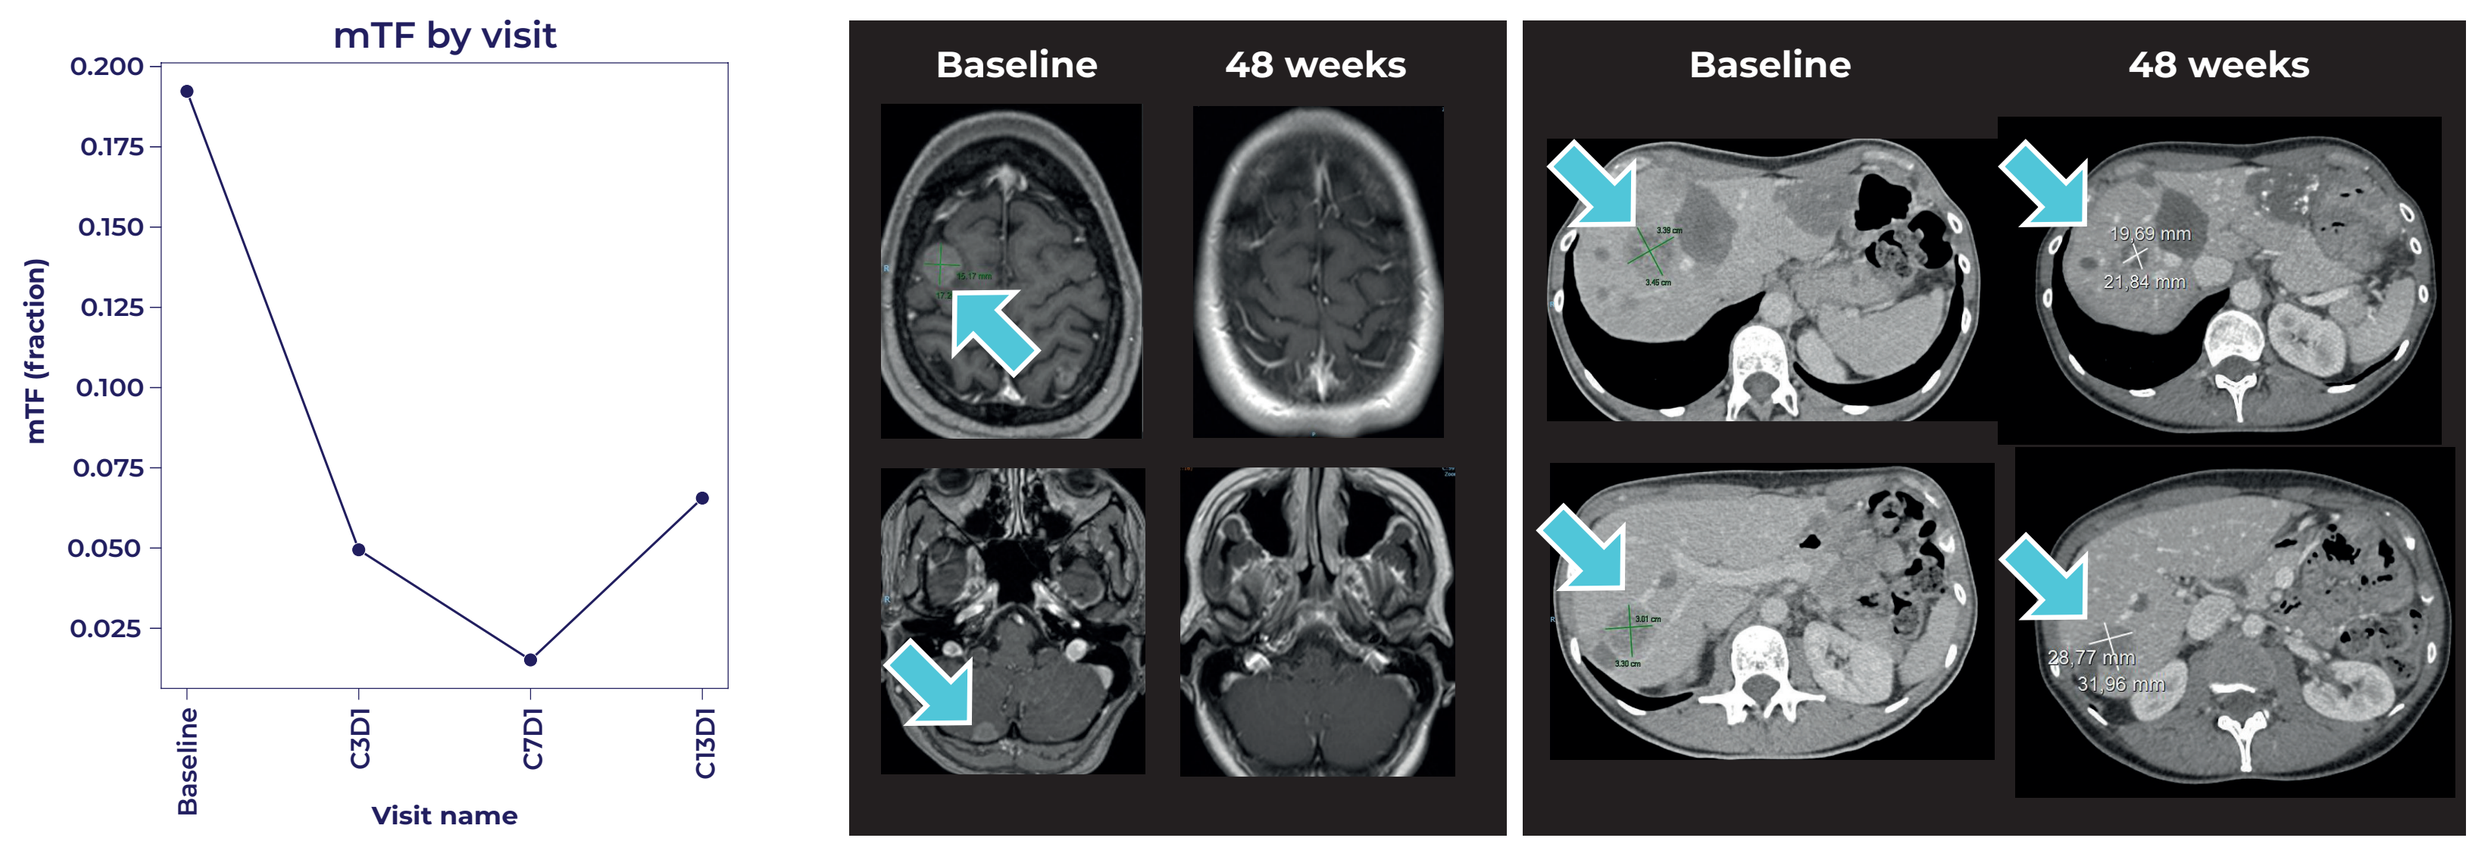

mTF by visit
mTF (fraction)
Baseline
C3D1
C7D1
C13D1
Visit name
Baseline
48 weeks
Baseline
48 weeks

Figure 5. 63 year-old female with KIF5B-RET fusion +ve NSCLC who previously received selpercatinib [NSCLC-4]

Patient history

- Prior therapy: Selpercatinib for 17 months (BOR PR) followed by platinum-based doublet chemotherapy + IO
- Patient treated with EP0031, 60mg QD; cPR
- ctDNA measurable at baseline, including KIF5B-RET fusion. Low methylation TF and ctDNA after treatment initiated, indicative of a molecular response (see adjacent figure)
- Patient was on treatment for 11 cycles / methylation TF and ctDNA remained low until C12D1 (EOT)

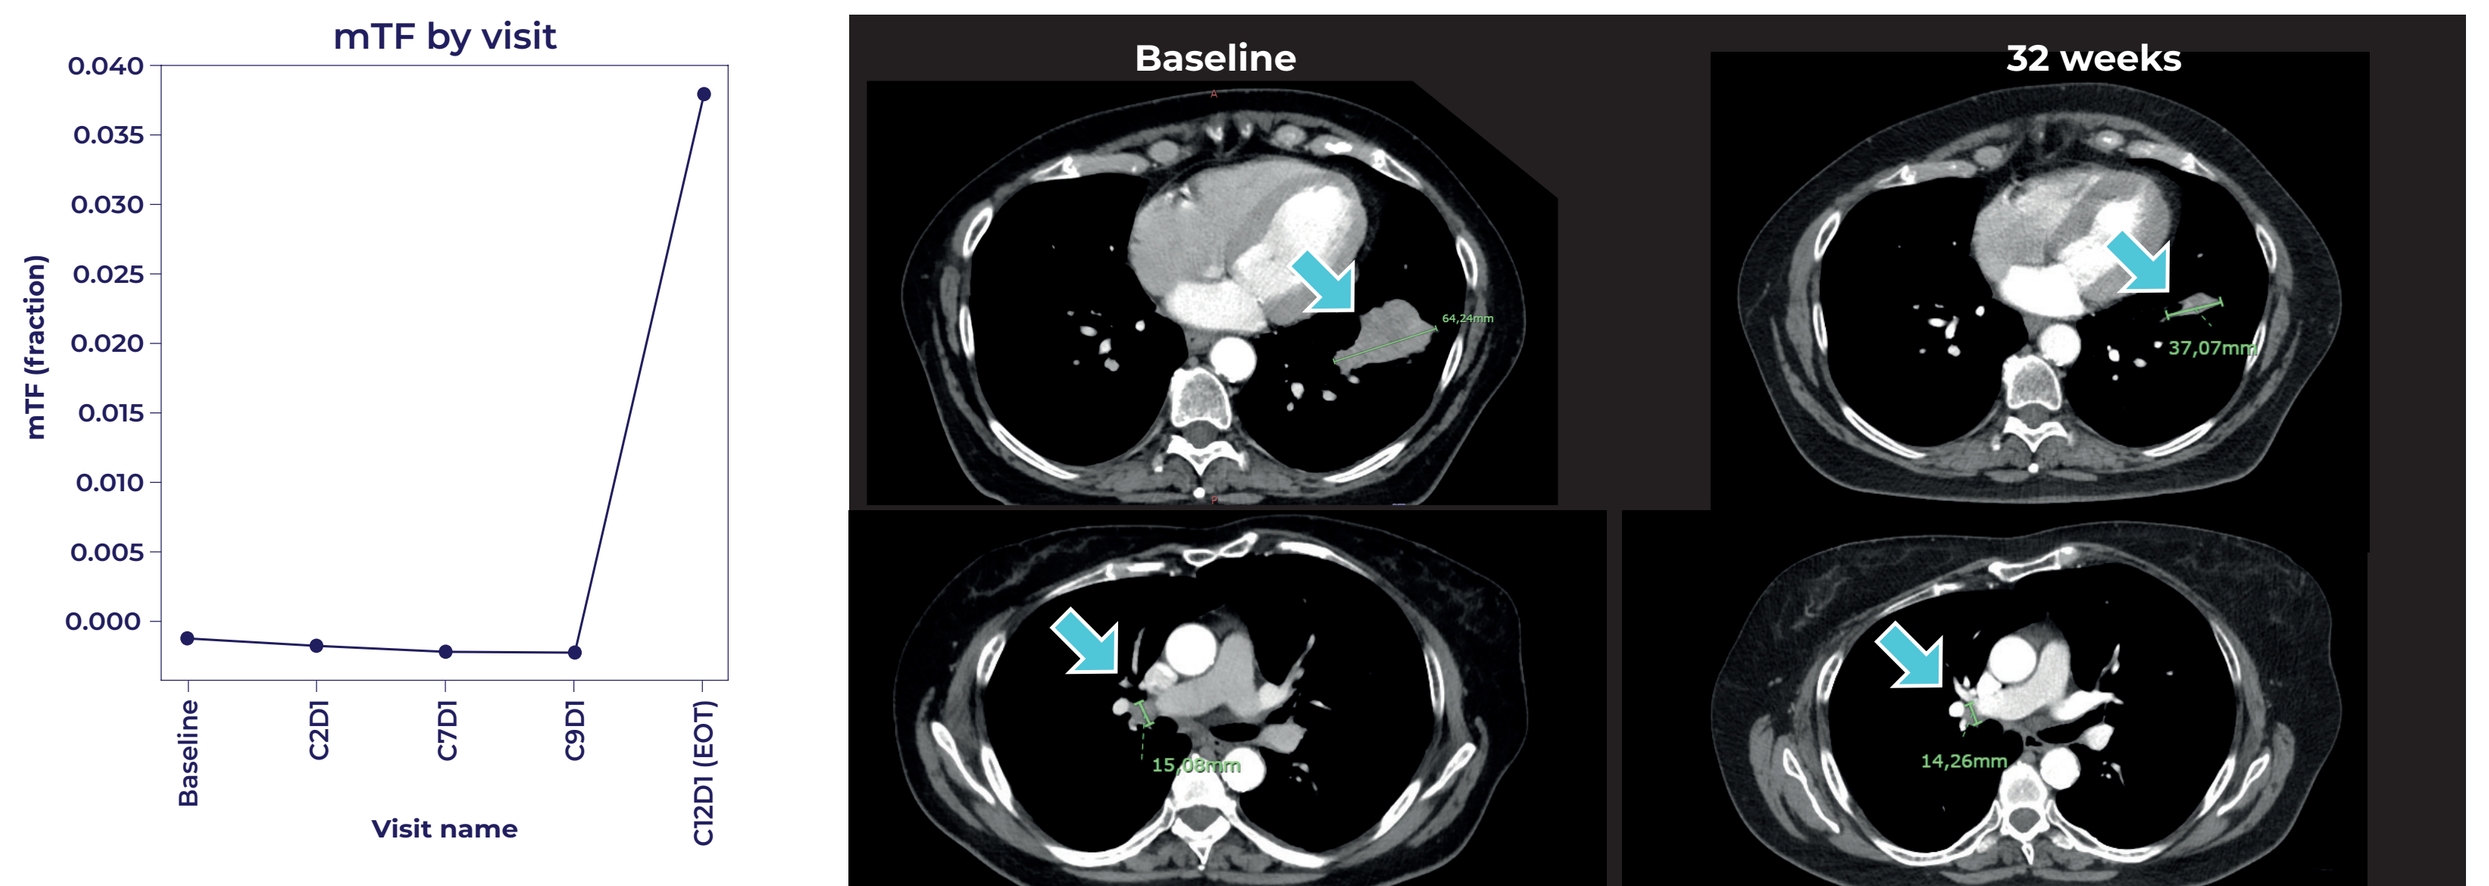

mTF by visit
mTF (fraction)
Baseline
C2D1
C7D1
C9D1
C12D1 (EOT)
Visit name
Baseline
32 weeks

Figure 6. 48 year-old female with KIF5B-RET fusion +ve NSCLC who previously received pralsetinib [NSCLC-9]

Patient history

- Prior therapy: 6 prior therapies including pralsetinib for 7 months (BOR PR) and platinum-based doublet chemotherapy + IO
- Patient treated with EP0031, 90mg QD; cPR; non-target lesions in the brain: complete resolution at C5
- ctDNA and methylation TF remained low until C11D1 (EOT) (see adjacent figure)

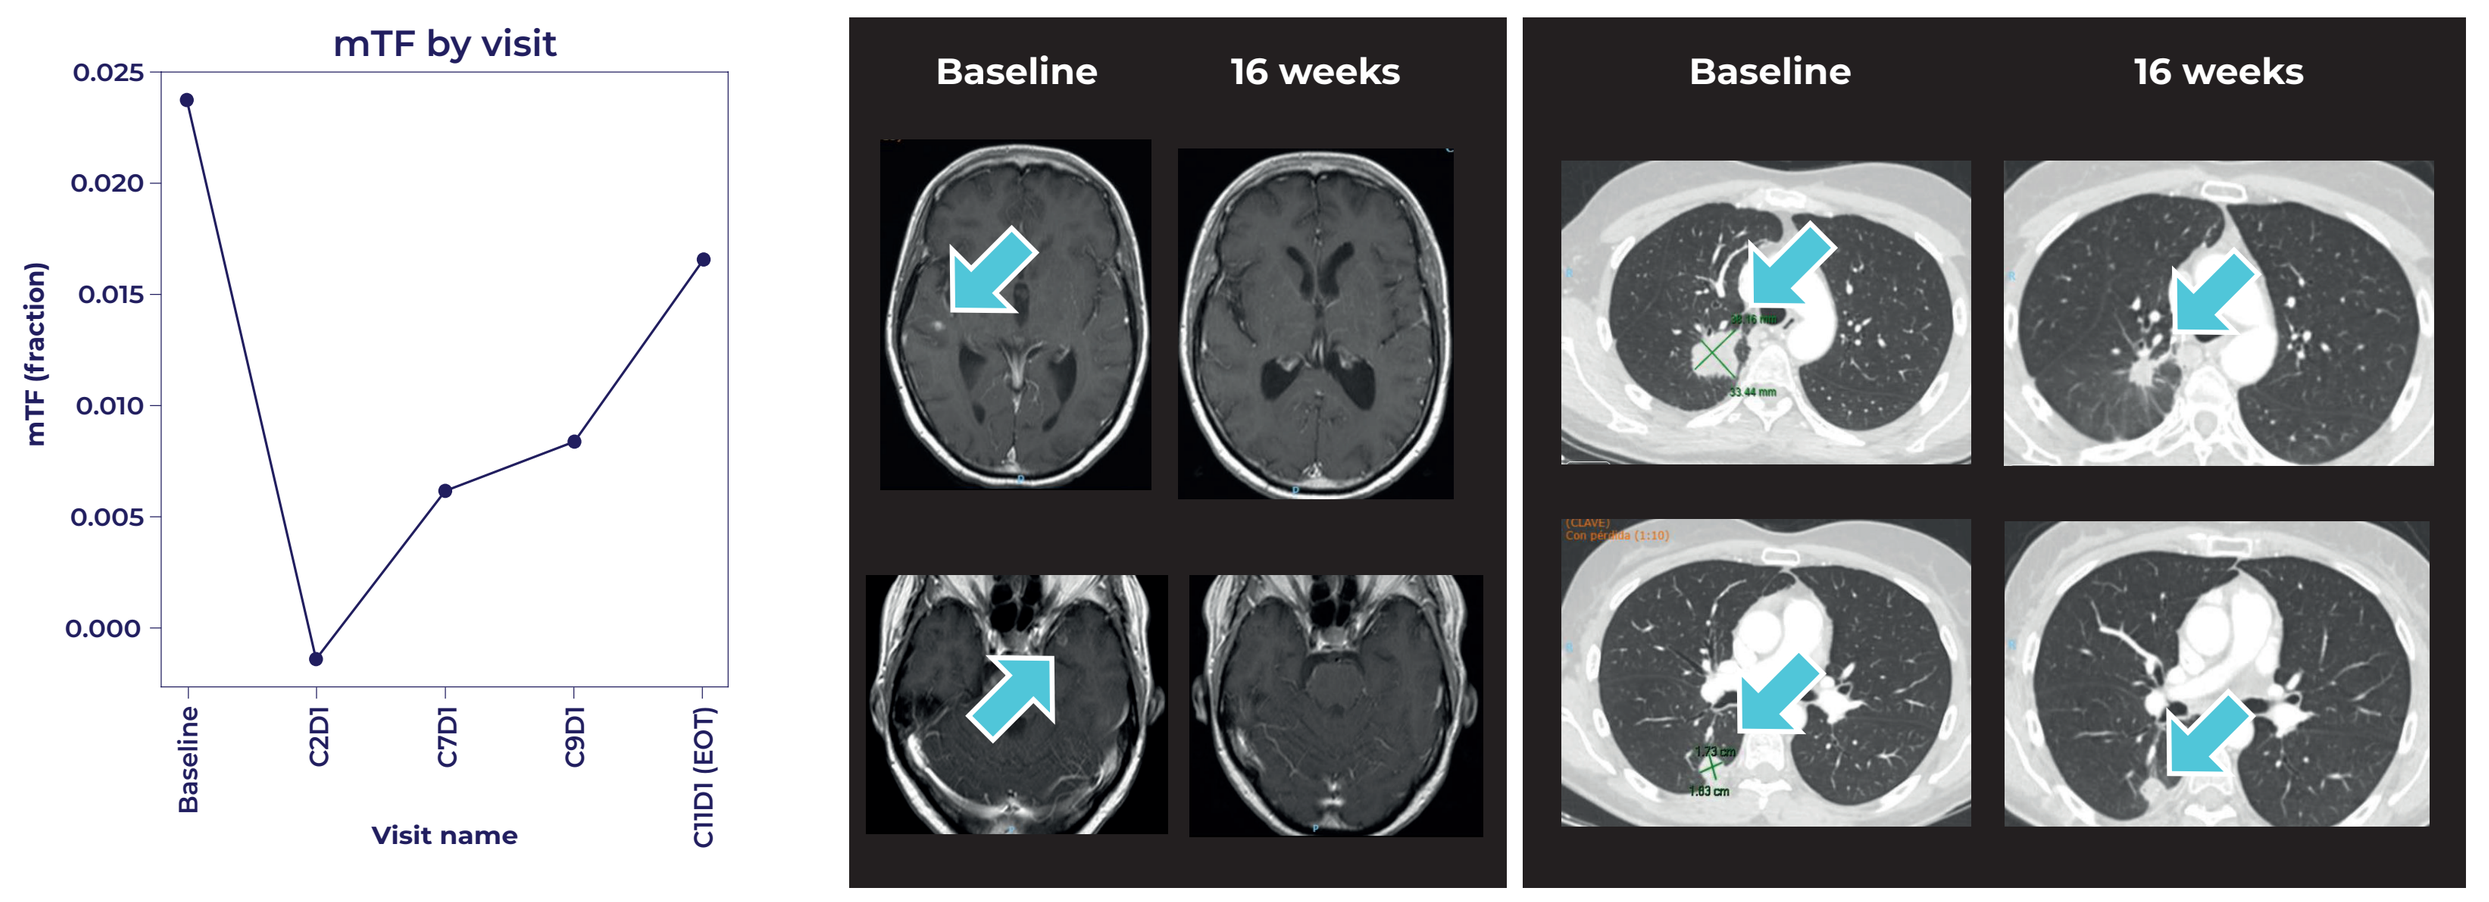

mTF by visit
mTF (fraction)
Baseline
C2D1
C7D1
C9D1
C11D1 (EOT)
Visit name
Baseline
16 weeks
Baseline
16 weeks

Figure 9. OncoPrint at baseline

Top 20 mutations for this cohort are shown
One NSCLC patient had no ctDNA samples analysed

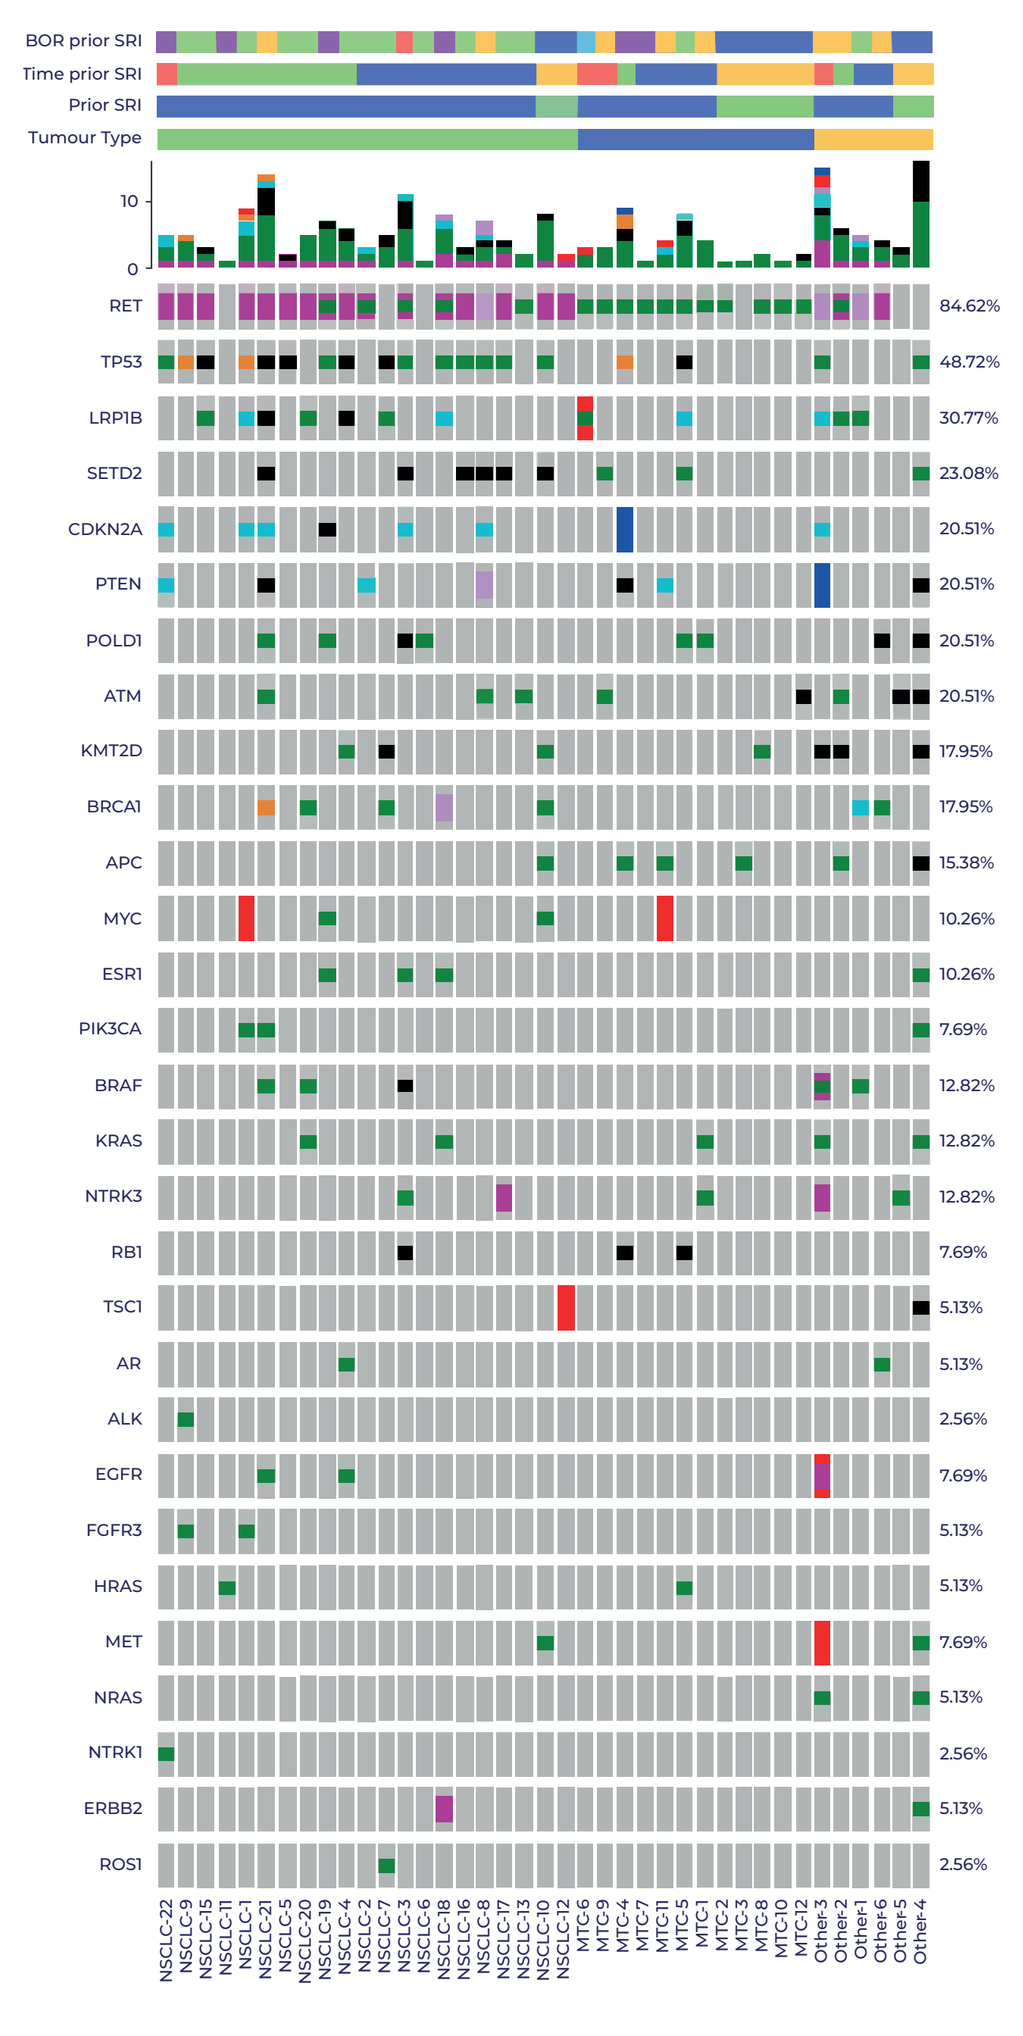

BOR prior SRI
Time prior SRI
Prior SRI
Tumour Type
RET 84.62%
TP53 48.72%
LRP1B 30.77%
SETD2 23.08%
CDKN2A 20.51%
PTEN 20.51%
POLD1 20.51%
ATM 20.51%
KMT2D 17.95%
BRCA1 17.95%
APC 15.38%
MYC 10.26%
ESR1 10.26%
PIK3CA 7.69%
BRAF 12.82%
KRAS 12.82%
NTRK3 12.82%
RB1 7.69%
TSC1 5.13%
AR 5.13%
ALK 2.56%
EGFR 7.69%
FGFR3 5.13%
HRAS 5.13%
MET 7.69%
NRAS 5.13%
NTRK1 2.56%
ERBB2 5.13%
ROS1 2.56%

- Most frequent baseline alterations alterations were RET fusions, followed by putative driver RET mutations; 1 patient had a RET structural variant
- NSCLC patients exhibited TP53 mutations more frequently than MTC and other solid tumour patients
- Other notable baseline mutations in NSCLC patients include LRPB1, SETD2 and CDKN2A (tumour suppressor genes)

Figure 10. OncoPrint at EOT

EOT somatic alterations are shown (non-pathogenic germline alterations not shown)

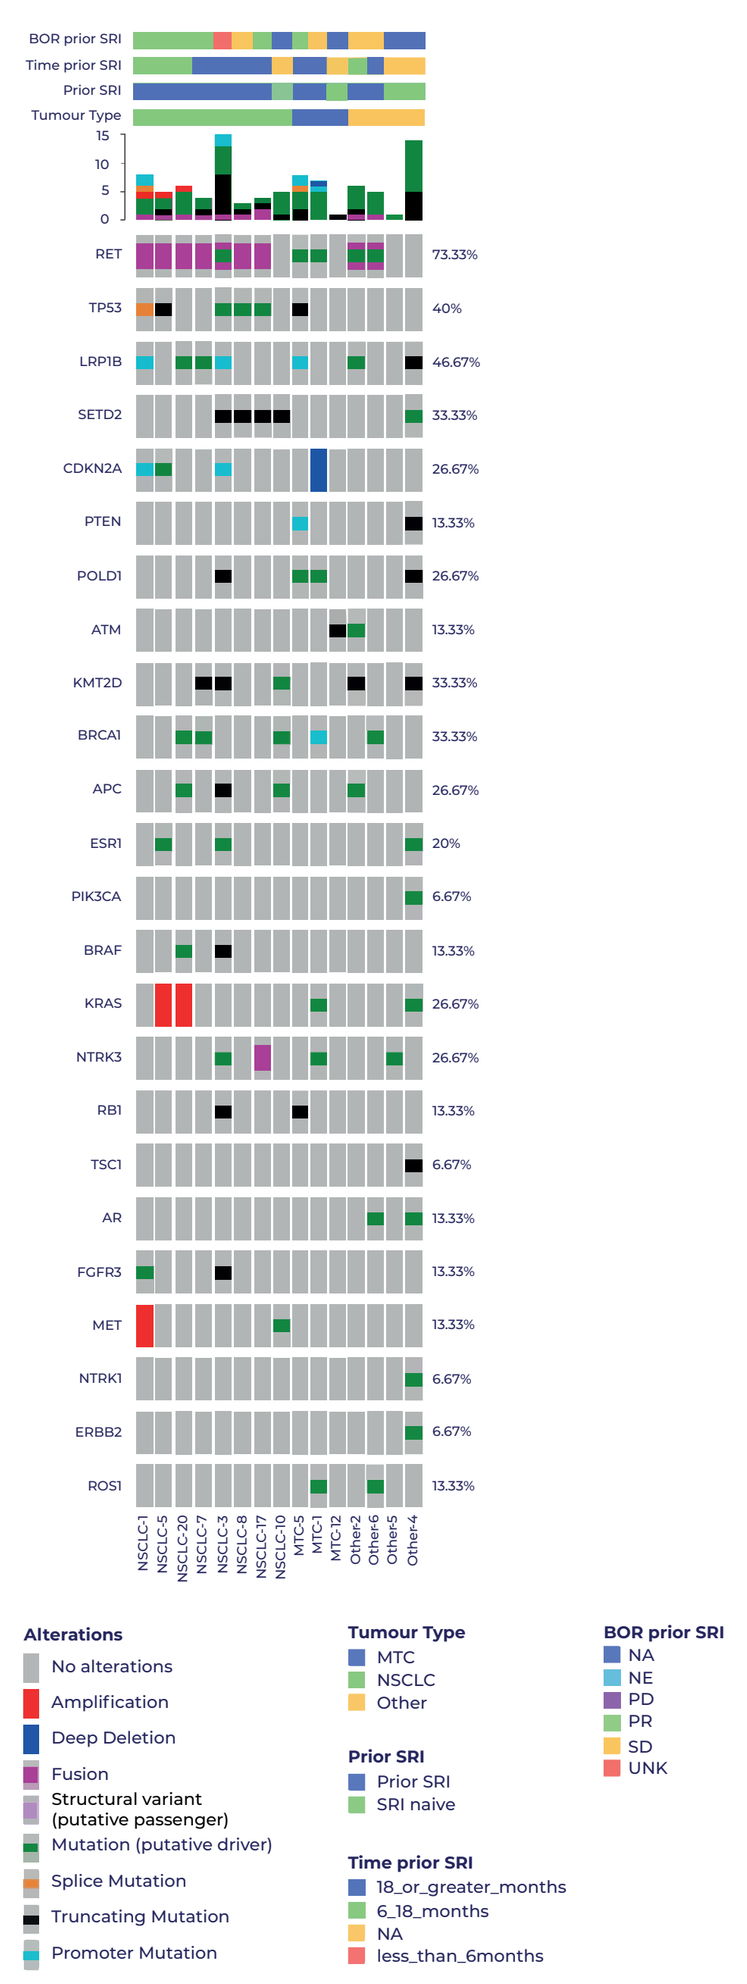

Alterations
No alterations
Amplification
Deep Deletion
Fusion
Structural variant (putative passenger)
Mutation (putative driver)
Splice Mutation
Truncating Mutation
Promoter Mutation
Tumour Type
MTC
NSCLC
Other
Prior SRI
Prior SRI
SRI naive
Time prior SRI
18_or_greater_months
6_18_months
NA
less_than_6months
BOR prior SRI
NA
NE
PD
PR
SD
UNK

- Most frequent mutations in the NSCLC EOT samples were off-target and included MET and KRAS amplifications
- On-target RET mutations were detected in 2 out of 5 MTC patients and 2 out of 6 other solid tumour patients

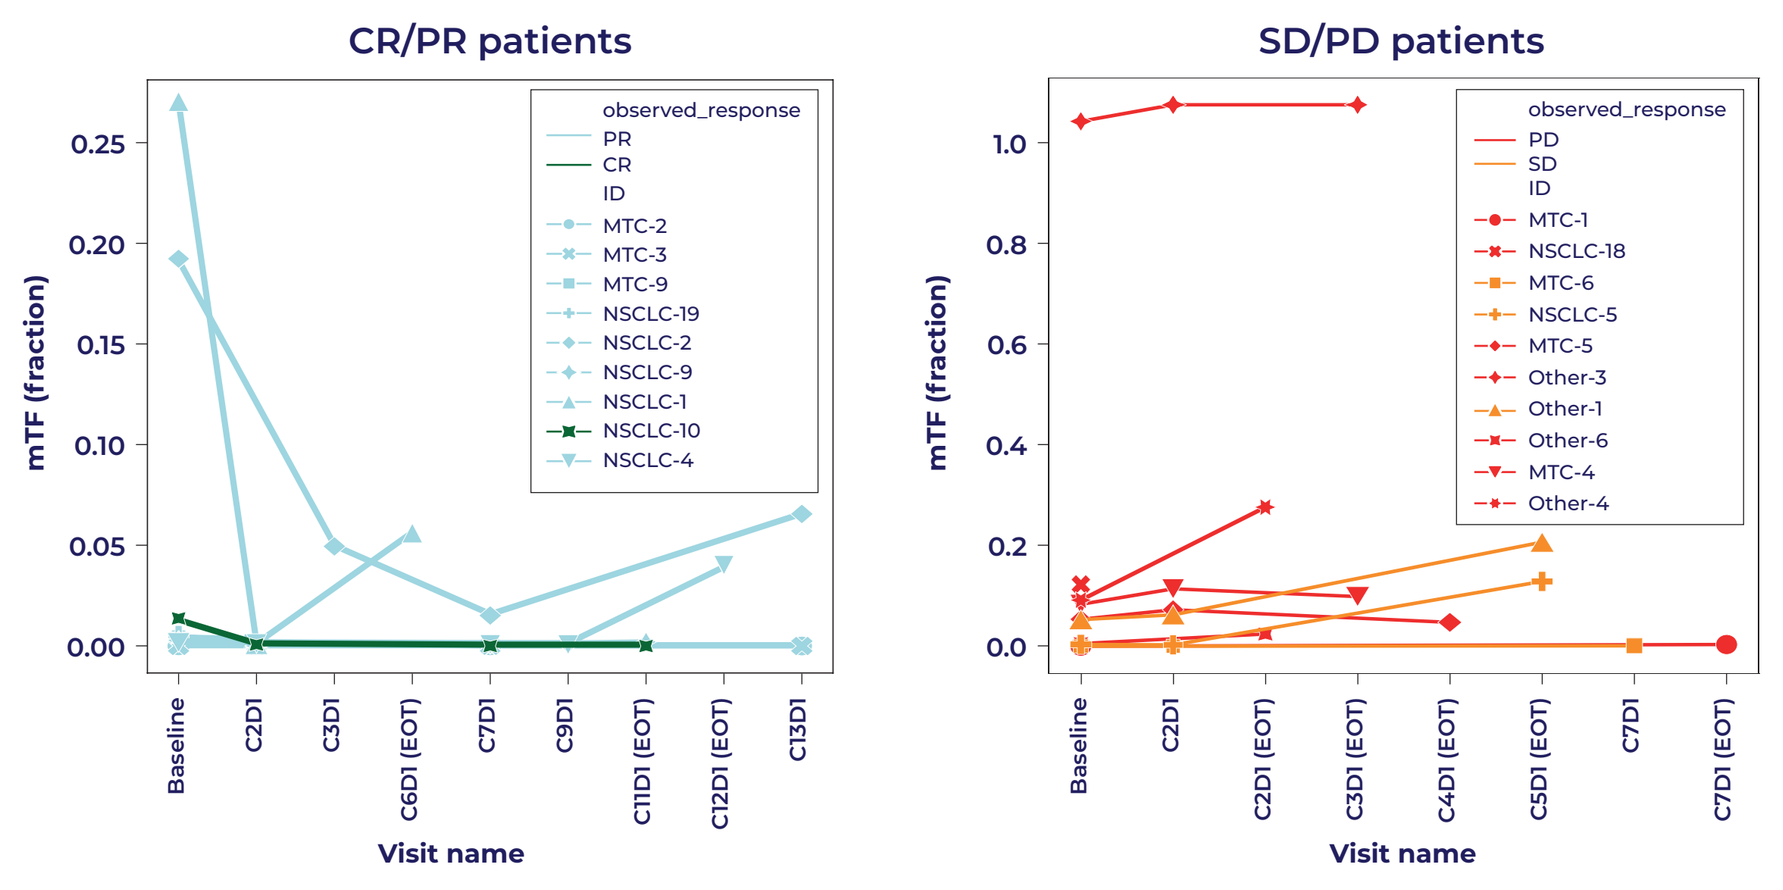

CR/PR patients
SD/PD patients
observed_response
mTF (fraction)
Visit name

Figure 7. Methylation tumour fraction (mTF) for all patients by visit and response

- Sustained reduction in methylation tumour fraction (mTF) in patients who achieve CR/PR (Left panel)
- Baseline mTF are higher in patients who have SD/PD as best response (Right panel)

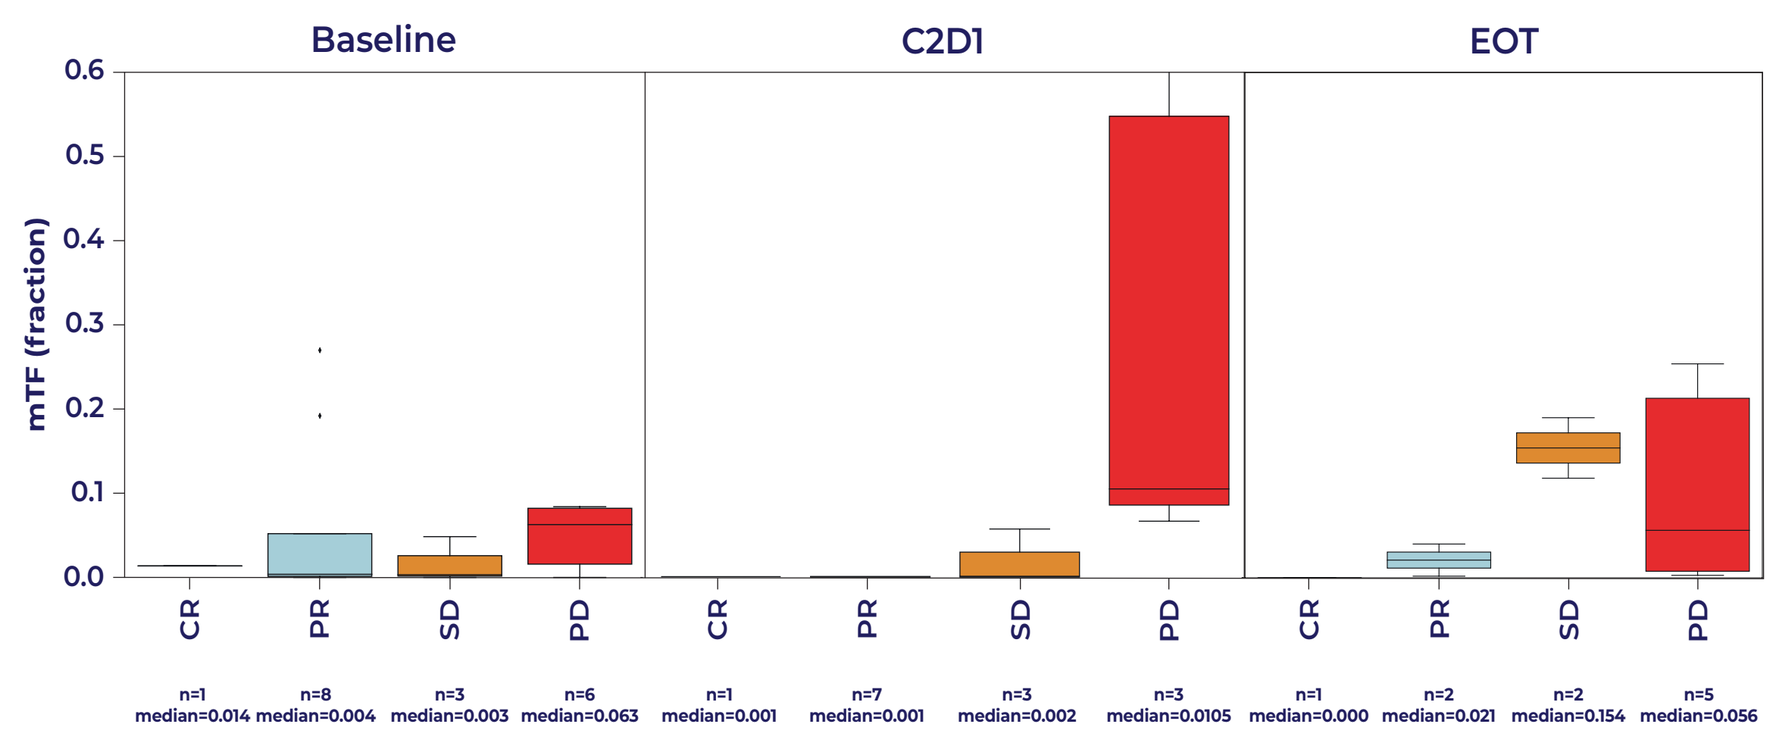

Baseline
C2D1
EOT
mTF (fraction)
CR n=1 median=0.014
PR n=8 median=0.004
SD n=3 median=0.003
PD n=6 median=0.063
CR n=1 median=0.001
PR n=7 median=0.001
SD n=3 median=0.002
PD n=3 median=0.0105
CR n=1 median=0.000
PR n=2 median=0.021
SD n=2 median=0.154
PD n=5 median=0.056

Figure 8. Grouped methylation TF by best overall response and visit

Box and whisker plot: the box covers the interquartile range (IQR) which contains 50% of the data; the line inside the box represents the median of the data; the whiskers extend to 1.5 times the IQR from the 1st quartile (lower edge of box) and 3rd quartile (upper edge of box). Any data points outside this range are considered outliers and are plotted as individual points

- Baseline (n=18) methylation TF are higher in patients who have PD as best response (Left panel)
- At C2D1 (n=14), reductions in methylation TF correlate with clinical response (middle panel); mTF consistently reduced in patients with CR or PR and minimal change in patients with SD
- At EOT (n=10), consistently low methylation TF for patients with CR or PR; significant increases in methylation TF compared to baseline for patients with SD and PD (Right panel)

SUMMARY & CONCLUSIONS

- Preclinical and clinical data indicates that EP0031 is a promising next gen SRI
- EP0031 has an encouraging safety and tolerability profile with evidence of efficacy in patients naïve to or previously treated with 1st gen SRIs, and activity against brain metastases
- Biomarker analysis support the efficacy of EP0031 in patients with prior SRI- independent of the presence of emergent RET resistance mutations - with tumours presenting with a high degree of heterogeneity:
 - At baseline, the most frequent RET alterations were RET fusions, followed by putative driver RET mutations; NSCLC patients also exhibited TP53 mutations more frequently than other patients; other notable baseline mutations in NSCLC patients include LRBP1, SETD2 and CDKN2A
 - ctDNA reductions in prior SRI treated patients included on-target RET resistance mutations (eg L730V/L730I)
 - At the end of EP0031 treatment, off-target alterations were most frequent in NSCLC patients regardless of length of time on treatment, whereas on-target RET mutations were seen in 4 out of 11 MTC / other solid tumour patients
 - Patients with low baseline methylation tumour fraction (mTF) were more likely to achieve CR/PR, whereas patients with higher methylation TF were more likely to have SD/PD as their best response
 - Low methylation TF at baseline and early reductions in ctDNA on treatment may be predictive of better outcomes with SRIs. Further evaluation is merited to assess whether methylation TF has potential as a routine method to assess treatment efficacy and treatment selection
- Multiple sites are active/opening in this Phase 2 study across USA, UK, Spain, France and UAE, and recruitment of additional countries/ sites continues

Acknowledgements

We are grateful to the patients and their loved ones for their key contribution to this work, and the participating sites for their hard work. We would also like to thank Guardant Health and A4P for their support across all biomarker research

References

1. Zhou Q et al, JCO 2023; 41 (suppl 16; abstract 3007)
2. Garralda E et al, JCO 2024; 42 (suppl 16; abstract 8556)
3. Jiang T et al, Cancer Res 2023; 83 (suppl 7; abstract 6601)
4. Chen S et al, Cancer Res 2023; 83 (suppl 7; abstract 3123)

DOI for Elena Garralda

Research: Novartis, Roche, Thermo Fisher, AstraZeneca, Taiho, BeiGene, Janssen
Consultant/Advisor: Roche, Ellipses Pharma, Boehringer Ingelheim, Janssen Global Services, Seattle Genetics, Thermo Fisher, MabDiscovery, Anaveon, Hengrui, Sanofi, Incyte, Medscape, Pfizer, Amgen
Speakers Bureau: Merck Sharp & Dohme, Roche, Thermo Fisher, Novartis, SeaGen
Employment: NEXT Oncology
Stocks: 1TRIALSP
Clinical Trials (Institutional): Multiple commercial sponsors

Study sponsored by Ellipses Pharma Limited

Presented at the ESMO Congress 2024, Barcelona, Spain. 13-17 September 2024.

For further information please contact Elena Garralda: egarralda@vhio.net Sonia Serrano: sonia@ellipses.life